INTERIM DATA ON DUAL INHIBITION OF POST- RADIOGENIC ANGIO-VASCULOGENESIS BY OLAPTESED PEGOL (NOX-A12) AND BEVACIZUMAB IN GLIOBLASTOMA FROM THE FIRST EXPANSION ARM OF THE PHASE 1/2 GLORIA TRIAL

Frank A. Giordano[1,2], Julian P. Layer[3,4], Sonia Leonardelli[4], Lea L. Friker[4,5], Christina Schaub[6], Thomas Zeyen[6], Elena Sperk[1,7], Peter Hambsch[8], Christoph Oster[9], Franziska Grau[3,10], Daniel Paech[10], Wolf Müller[11], Torsten Pietsch[5], L. Christopher Schmeel[3], Oliver Grauer[12], Mirjam Renovanz[13], Ghazaleh Tabatabai[13], Sied Kebir[10], Sotirios Bisdas[14], Erdem Güresir[15], Martin Glas[10], Clemens Seidel[11], Michael Hölzel[4] and Ulrich Herrlinger[6]

[1] Department of Radiation Oncology, University Medical Center Mannheim, University of Heidelberg; [2] DKFZ-Hector Cancer Institute at the University Medical Center Mannheim; [3] Department of Radiation Oncology, University Hospital Bonn, University of Bonn; [4] Institute of Experimental Oncology, University Hospital Bonn, University of Bonn; [5] Institute of Neuropathology, University Hospital Bonn, University of Bonn; [6] Division of Clinical Neuro-Oncology, Department of Neurology, University Hospital Bonn; [7] Clinical Trials Unit, University Medical Center Mannheim, University of Heidelberg; [8] Department of Radiation Oncology, University Hospital Leipzig, University of Leipzig; [9] Division of Clinical Neurooncology, Department of Neurology, University Hospital Essen, University Duisburg-Essen; [10] Department of Radiation Oncology, University Hospital Bonn, University of Bonn; [11] Institute of Neuropathology, University Hospital Leipzig, University of Leipzig; [12] Department of Neurology, University Hospital Münster, University of Münster; [13] Department of Neurology, University Hospital Tübingen, University of Tübingen; [14] Lysholm Department of Neuroradiology, National Hospital for Neurology and Neurosurgery, London; [15] Department of Neurosurgery, University Hospital Leipzig, University of Leipzig

SNO Annual Meeting 2023
CTNI-62

1 Rationale

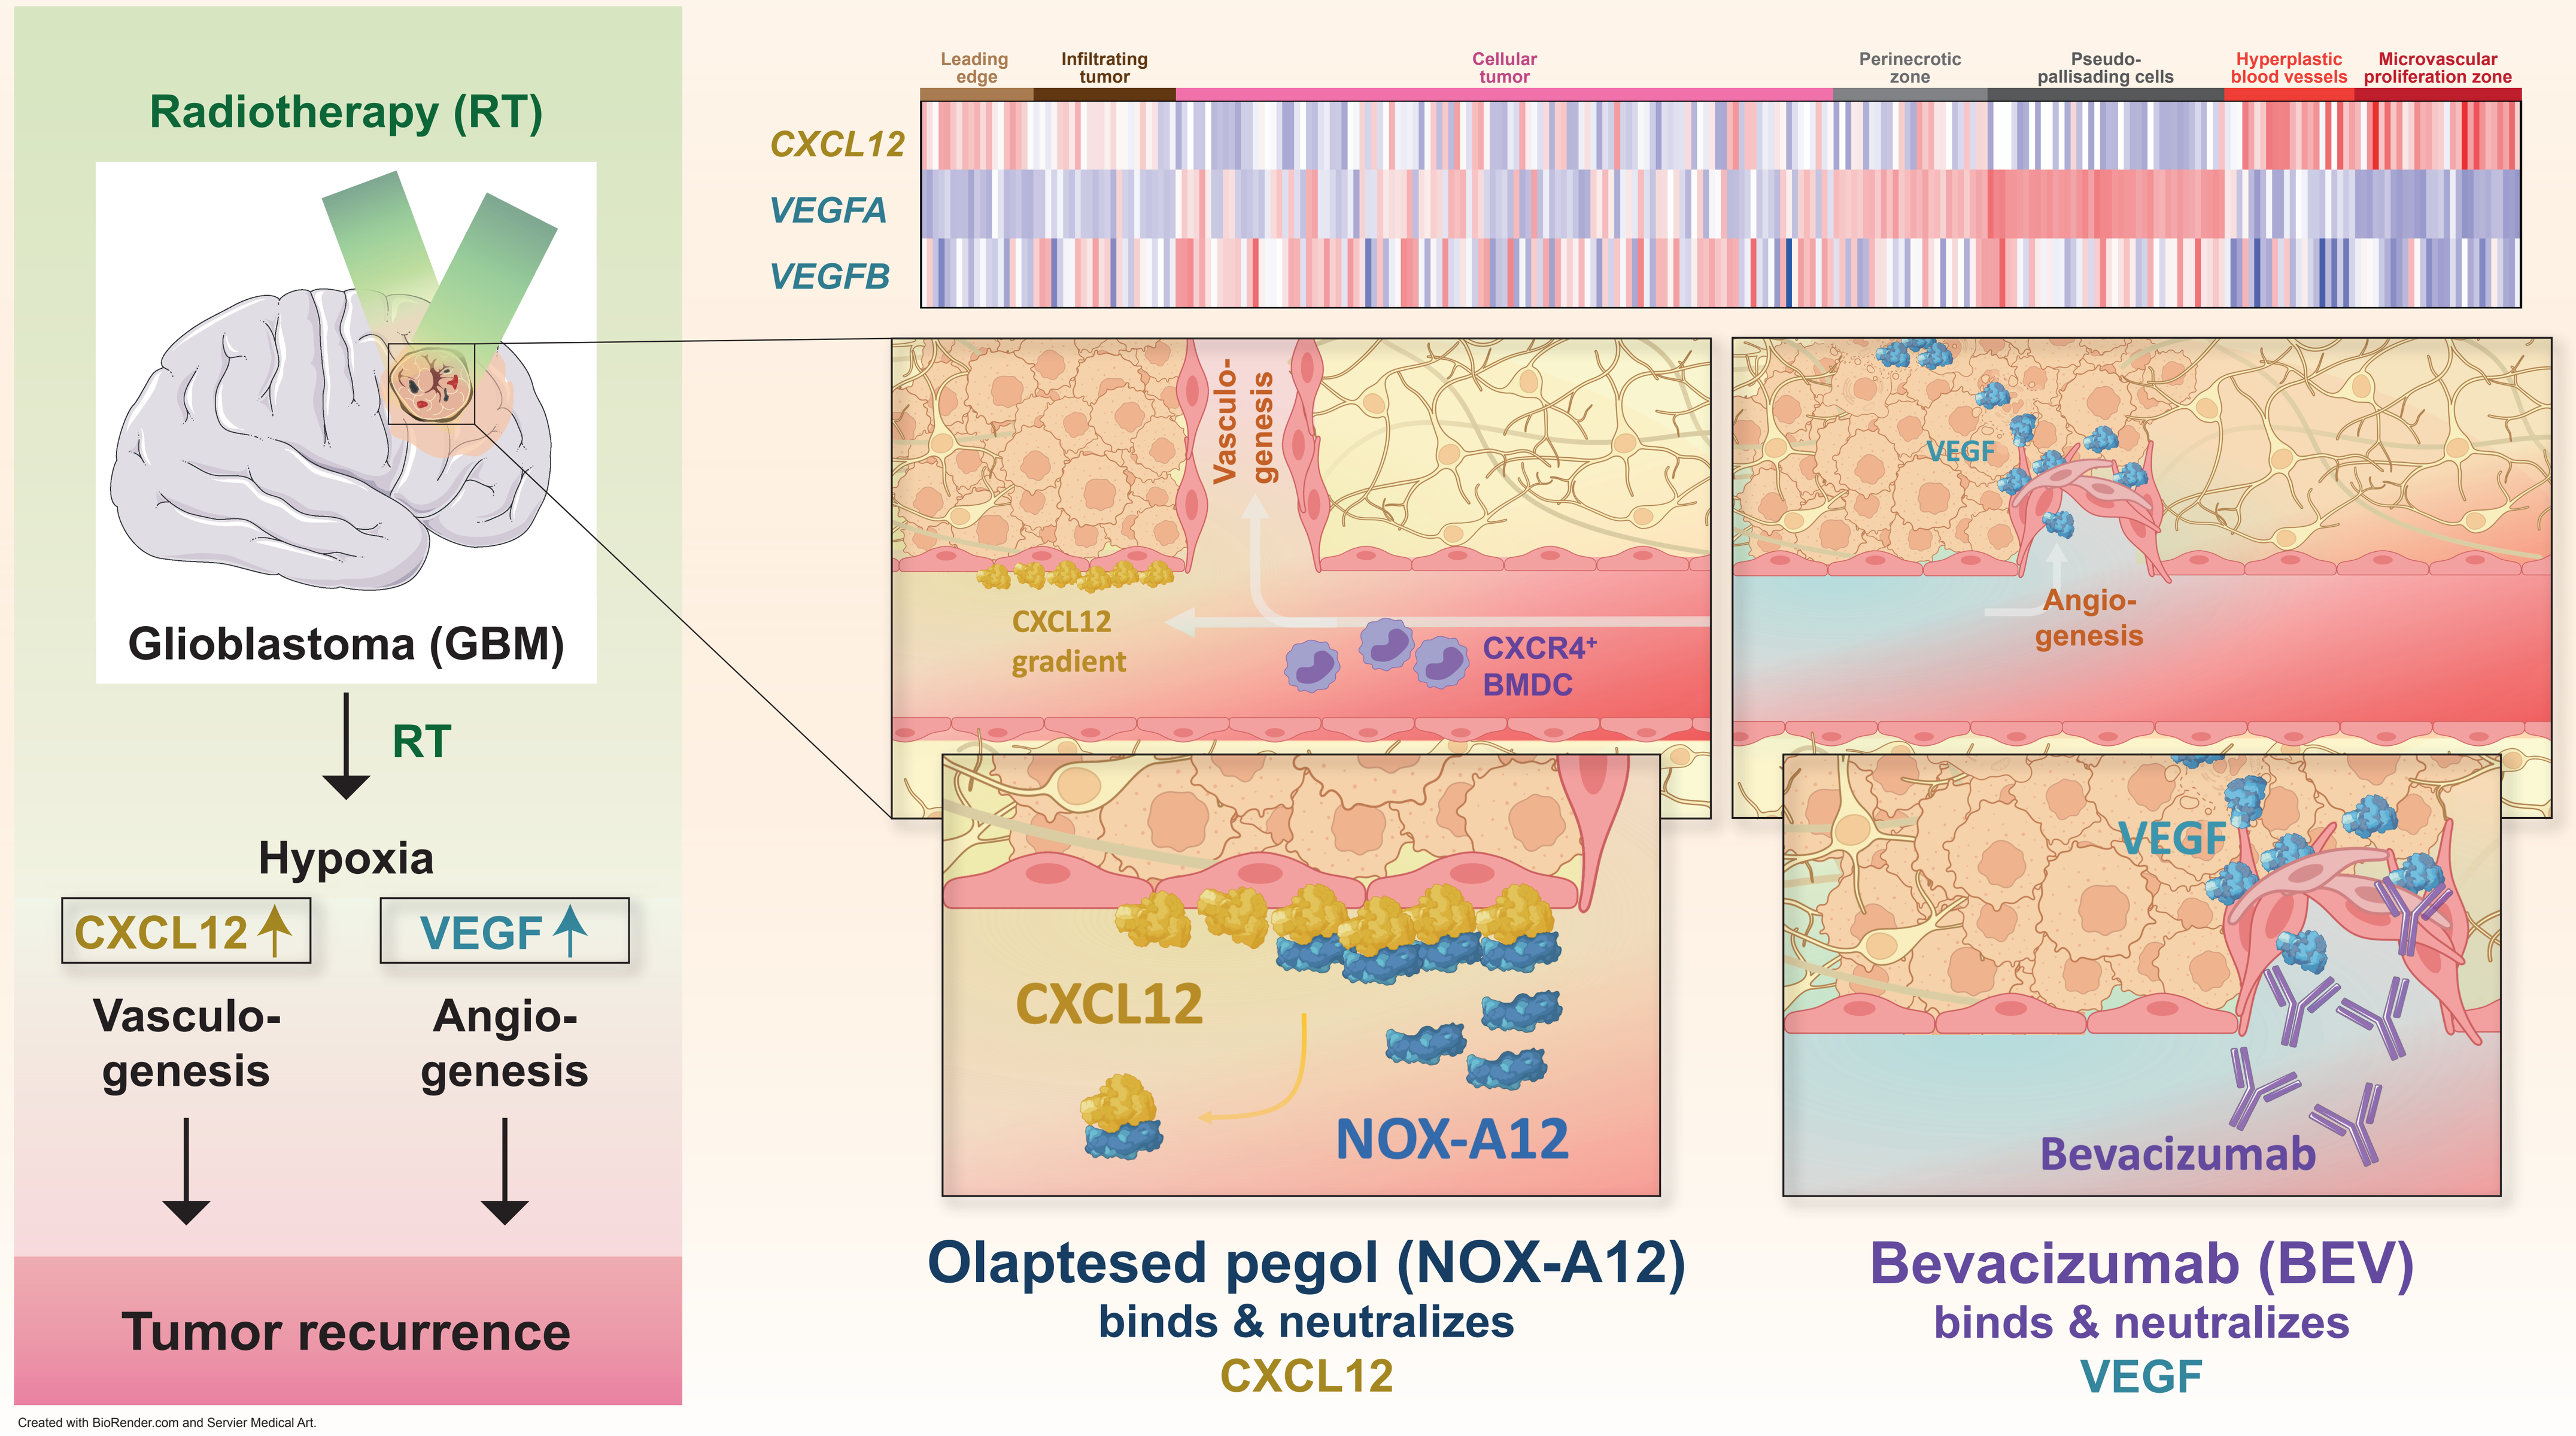
Radiotherapy (RT)
Glioblastoma (GBM)
RT
Hypoxia
CXCL12
VEGF
Vasculo-genesis
Angio-genesis
Tumor recurrence
Leading edge
Infiltrating tumor
Cellular tumor
Perinecrotic zone
Pseudo-pallisading cells
Hyperplastic blood vessels
Microvascular proliferation zone
CXCL12
VEGFA
VEGFB
Vasculo-genesis
CXCL12 gradient
CXCR4+ BMDC
VEGF
Angio-genesis
CXCL12
NOX-A12
VEGF
Bevacizumab

Created with BioRender.com and Servier Medical Art.

Olaptesed pegol (NOX-A12)
binds & neutralizes
CXCL12

Bevacizumab (BEV)
binds & neutralizes
VEGF

2 Study design

Inclusion criteria
- Newly-diagnosed supratentorial GBM (WHO CNS grade 4)
- MGMT promoter unmethylated
- Incomplete resection
- ECOG ≤ 2

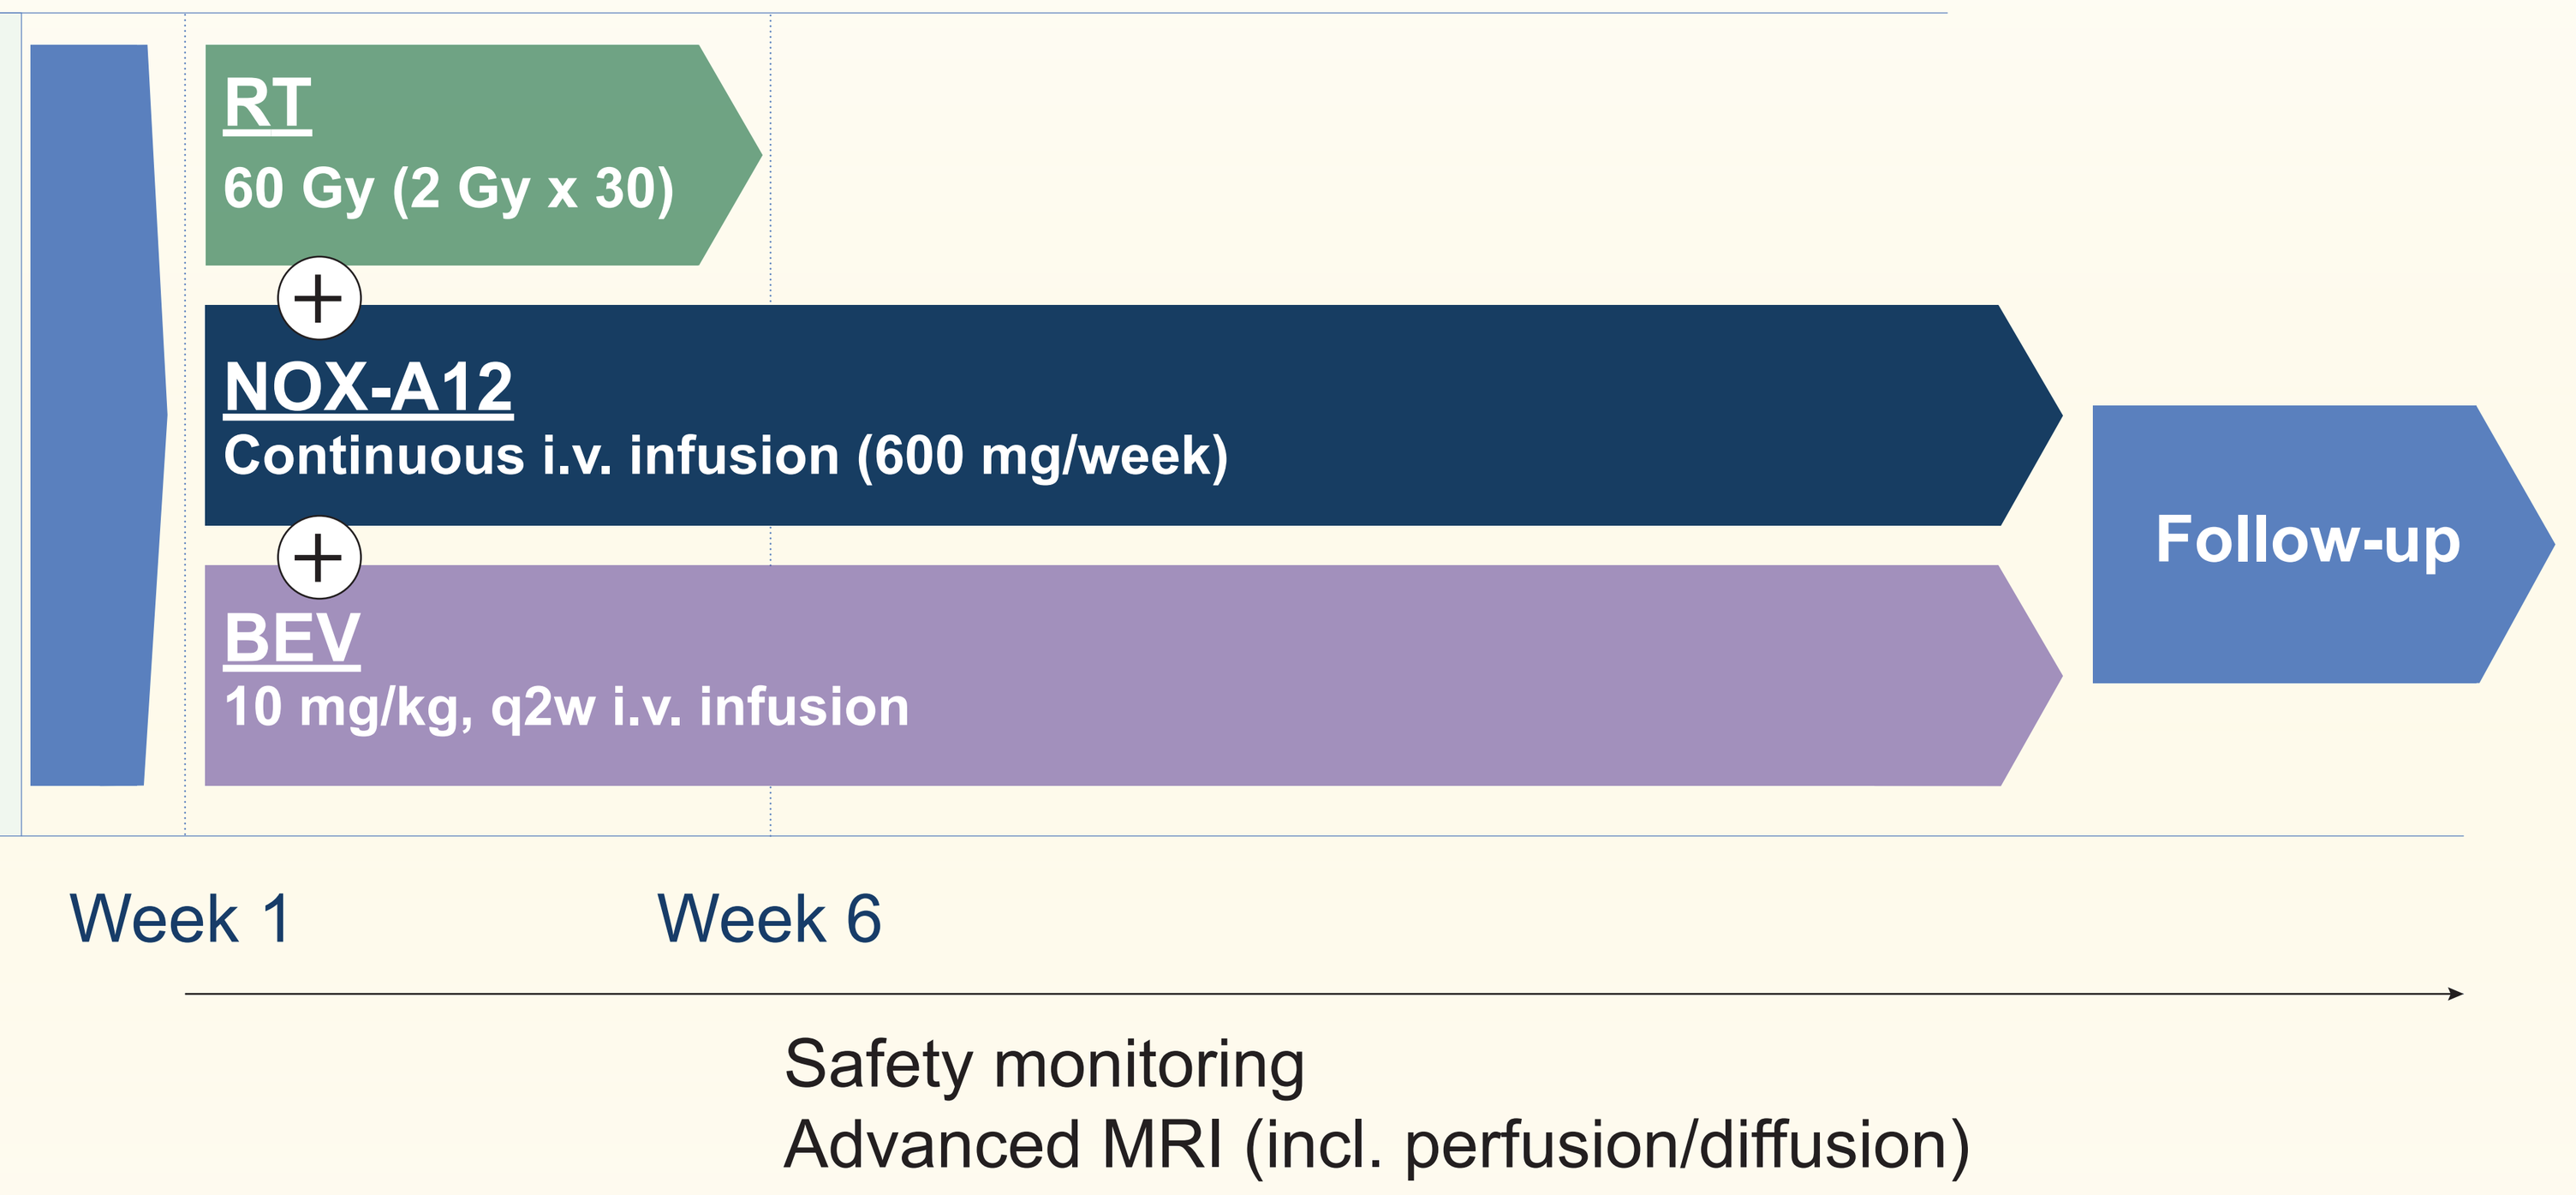
RT
60 Gy (2 Gy x 30)
NOX-A12
Continuous i.v. infusion (600 mg/week)
BEV
10 mg/kg, q2w i.v. infusion
Follow-up
Week 1
Week 6
Safety monitoring
Advanced MRI (incl. perfusion/diffusion)

Primary endpoint:	Safety (incidence of adverse events)
Secondary endpoint:	NOX-A12 plasma levels, radiographic response, PFS, OS
Exploratory endpoints:	Multiplexed immunofluorescence imaging (CODEX®) - assessment of tissue biomarkers

- 6 patients with newly diagnosed incompletely resected GBM with ECOG≤2, age≥18 and without MGMT promoter methylation were enrolled
- All patients received standard RT (60 Gy in 30 fractions) plus continuous (24/7) i.v. infusions of NOX-A12 and bevacizumab q2w

3 Safety

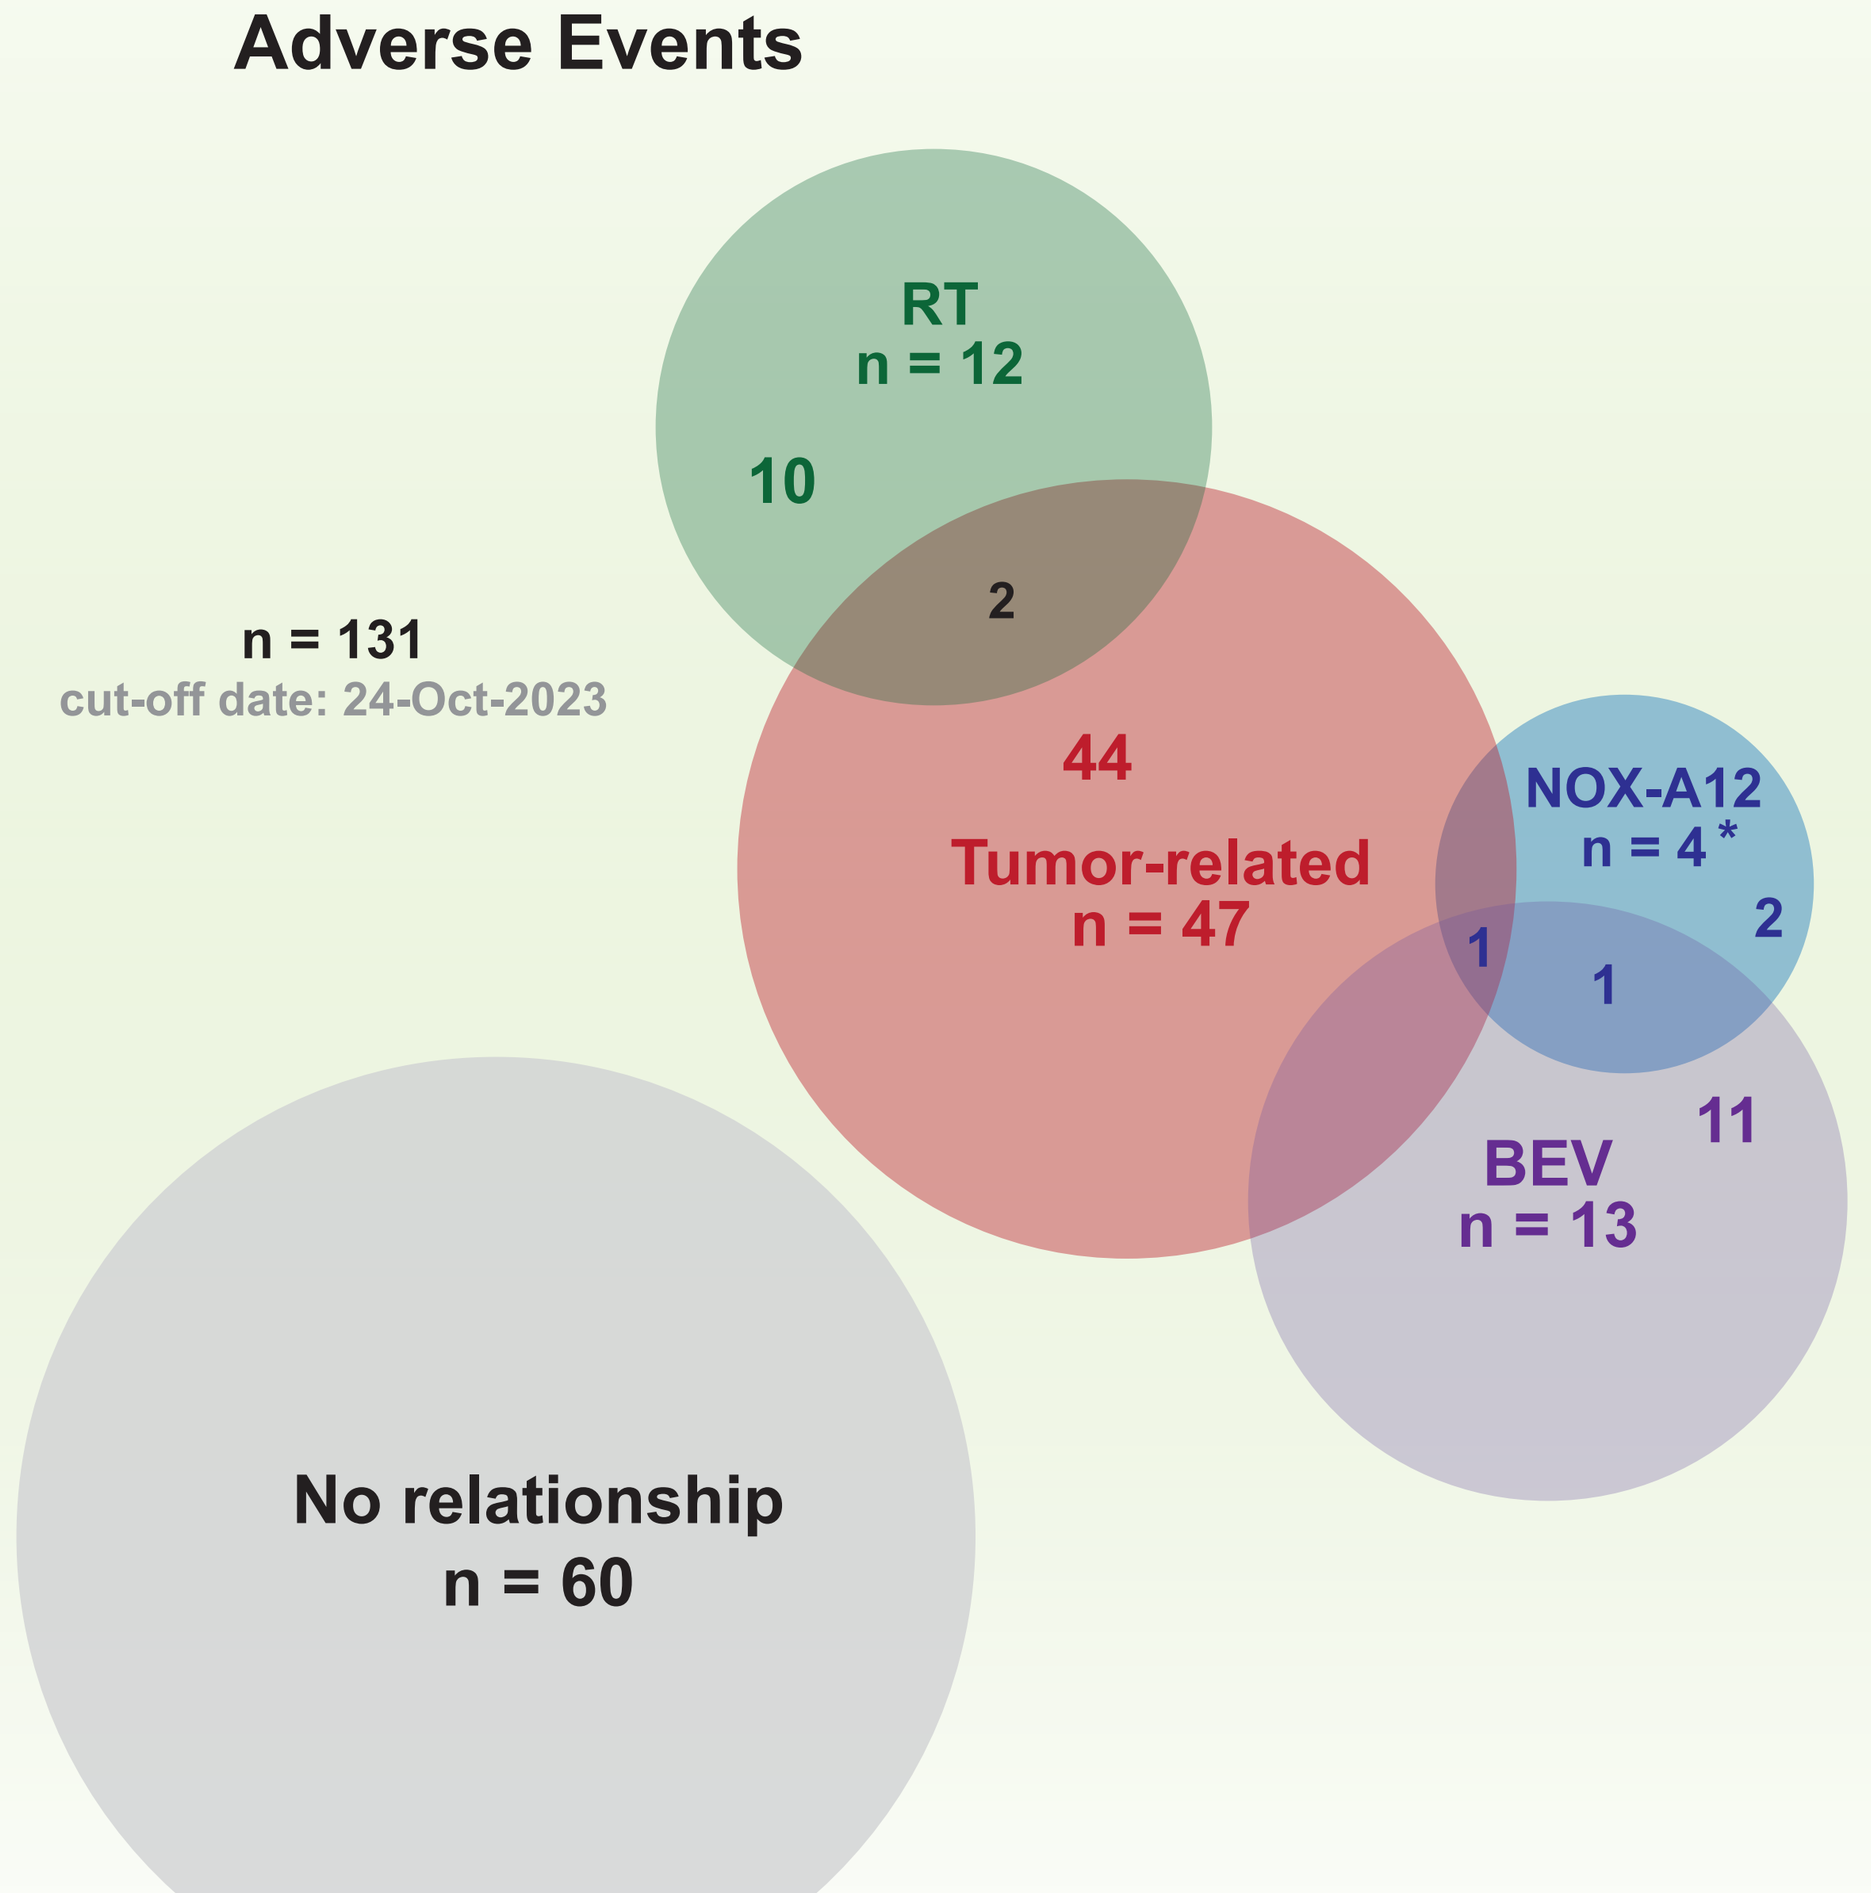
Adverse Events
n = 131
cut-off date: 24-Oct-2023
RT
n = 12
10
2
44
Tumor-related
n = 47
NOX-A12
n = 4*
2
1
1
11
BEV
n = 13
No relationship
n = 60

- Treatment was well-tolerated and safe
- No dose-limiting toxicities
- No treatment-related deaths
- No unexpectedly high toxicity of bevacizumab
- Of all G≥2 adverse events (n=88), 4 G2 events (4.5%) were considered to be (also) NOX-A12-related

***NOX-A12-related**

2x infusion site reaction G2
1x fatigue G2 (also related to GBM and BEV)
1x edema G2 (also related to BEV)

4 Radiographic response

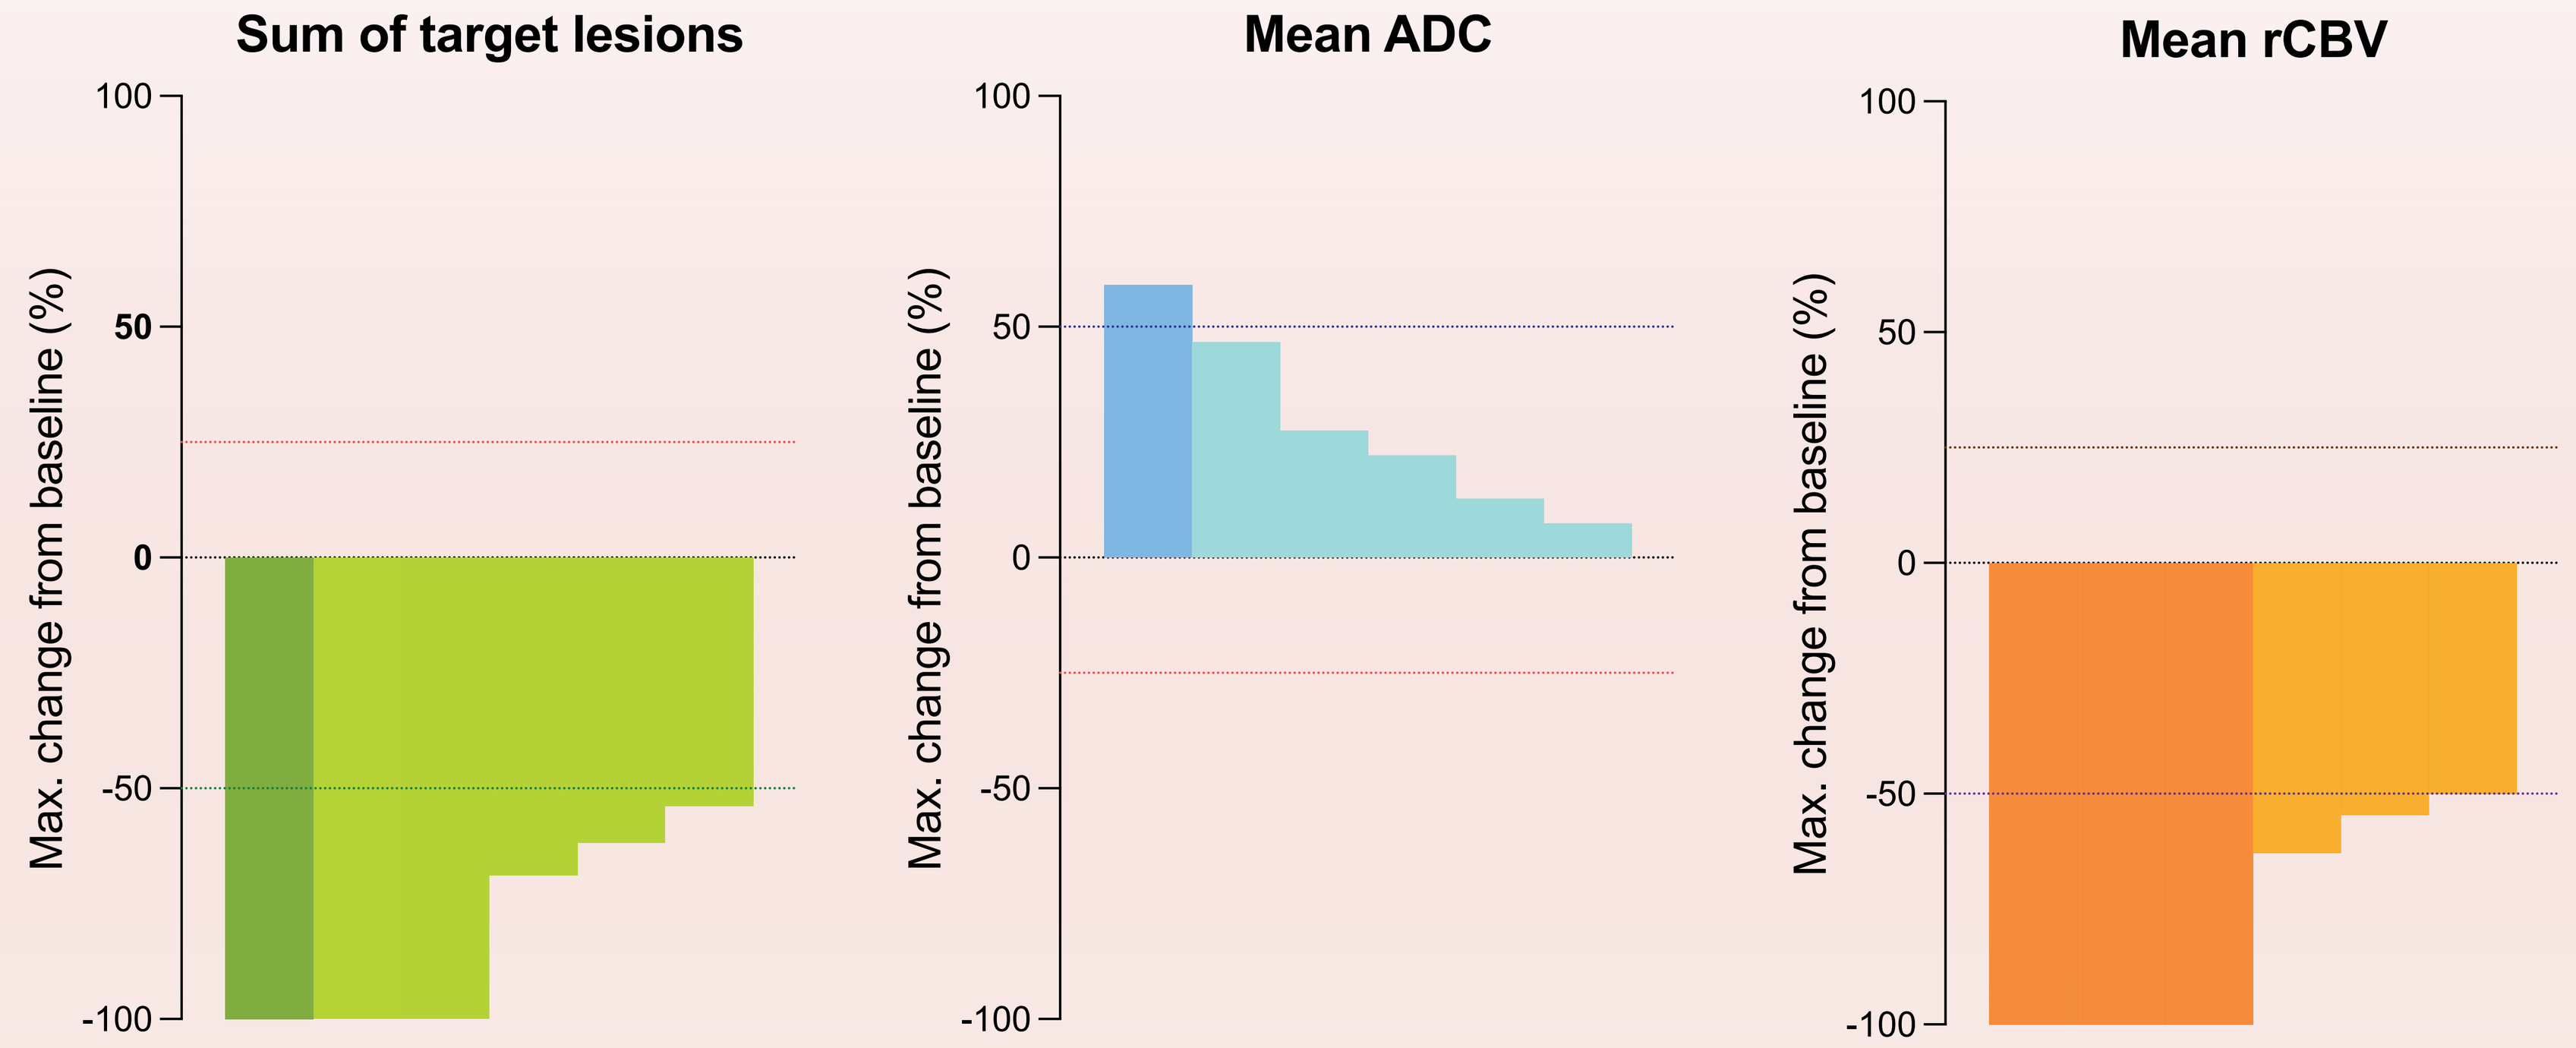
Sum of target lesions
Mean ADC
Mean rCBV
Max. change from baseline (%)
100
50
0
-50
-100

- With an overall response rate (ORR) of 83.3% under treatment, one patient achieved confirmed complete response (CR), four patients partial response (PR) and one patient stable disease (SD) as per mRANO
- The median best response under treatment was -68.9% (-53.8% to -100%) for target lesion sums and -100% (-99.9% to -100%) for non-target lesion (NTL) sums
- In all three patients with NTLs ≥ one lesion disappeared
- The median best change from baseline of the apparent diffusion coefficient (ADC) was 24.9% (7.3% to 59.1%) and the median best reduction of the highly perfused tumor fraction was 98.2% (61.3% to 100%).

Radiotherapy + NOX-A12 + bevacizumab in chemotherapy refractory **GBM**
Safe I No DLT I Encouraging improved efficacy of dual inhibition of angio-vasculogenesis
Treatment and follow-up ongoing

5 Clinical outcome

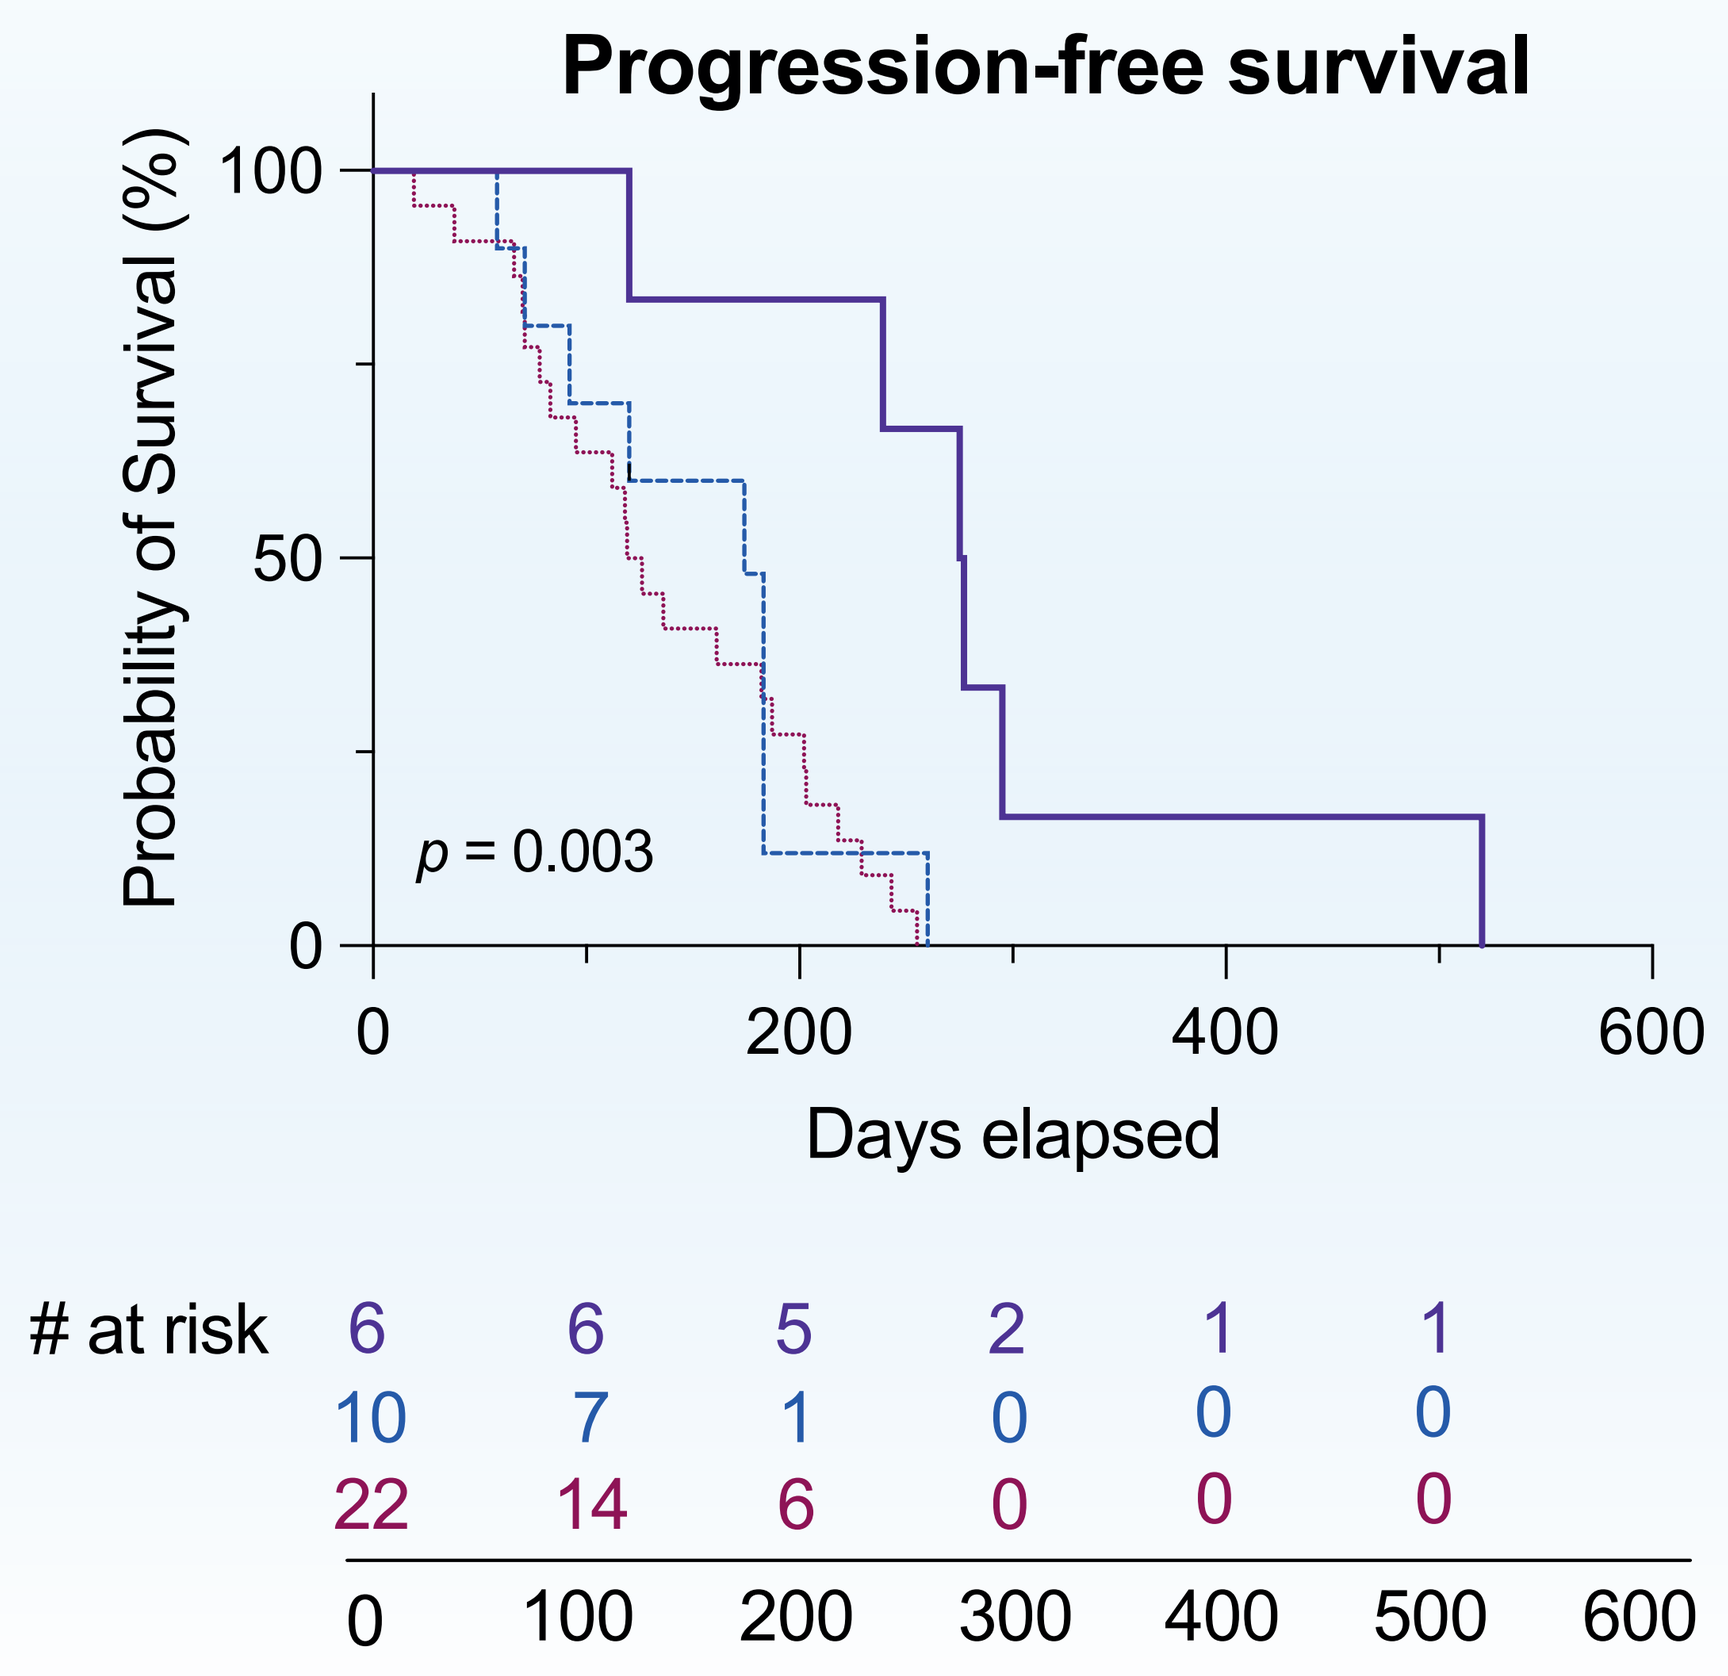
Progression-free survival
Probability of Survival (%)
100
50
0
p = 0.003
0
200
400
600
Days elapsed
at risk
6 6 5 2 1 1
10 7 1 0 0 0
22 14 6 0 0 0
0 100 200 300 400 500 600

	median PFS	median OS	
SOC	123 days	289 days	SOC
RT+NOX-A12	174 days	389 days	RT+NOX-A12
RT+NOX-A12+BEV	276 days	not reached	RT+NOX-A12+BEV

- At a median follow-up of 18.3 months, three patients deceased and median overall survival (OS) is not reached
- 67% landmark OS at 18 months
- Early death of one patient after 3.9 months due to distant leptomeningeal spread
- Significantly improved progression- free (PFS) and overall survival under triple treatment with RT + NOX-A12 + BEV compared to matched standard of care (SOC) cohort
- Treatment and follow-up are ongoing

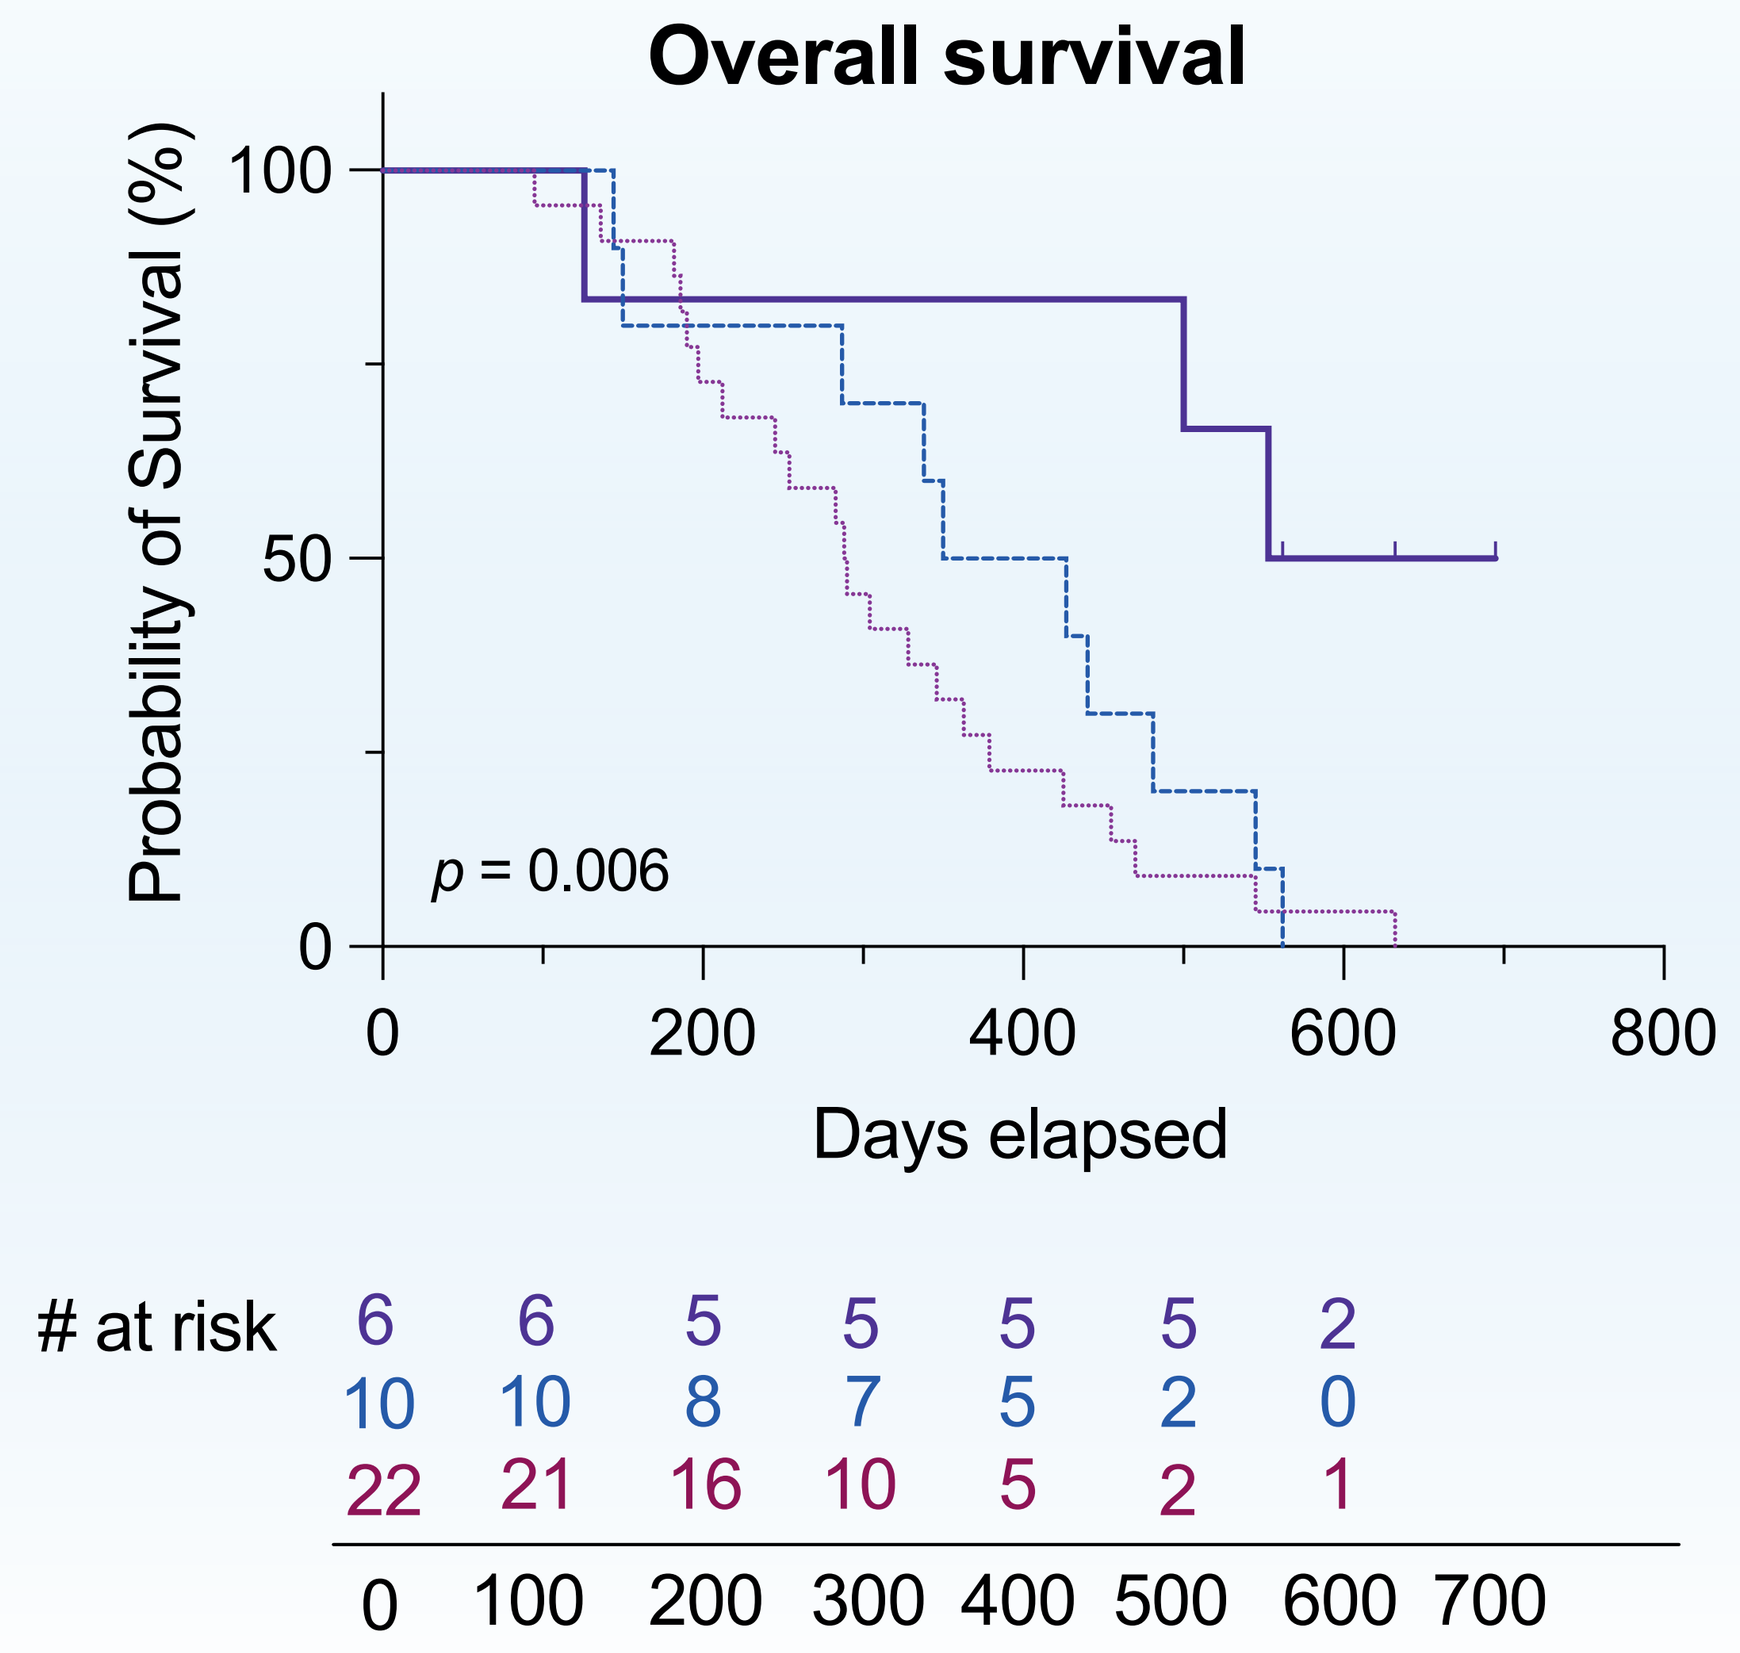
Overall survival
Probability of Survival (%)
100
50
0
p = 0.006
0
200
400
600
800
Days elapsed
at risk
6 6 5 5 5 5 2
10 10 8 7 5 2 0
22 21 16 10 5 2 1
0 100 200 300 400 500 600 700

Trial sponsored by TME Pharma AG. Registered with clinicaltrials.gov, ID: NCT04121455, and EudraCT 2018-004064-62

Frank.Giordano@umm.de

Copies of this poster are for personal use only and may not be reproduced without permission from the authors.